Ilofotase alfa exerts renal protective effects in patients with sepsis-associated acute kidney injury

Kathleen Liu[1], Kent Doi[2], Rinaldo Bellomo[3], Juliane Bernholz[4], Maarten Kraan[4], and Peter Pickkers[5] on behalf of the trial steering committee

[1] Division of Nephrology and Critical Care Medicine, University of California, San Francisco, United States; [2] Emergency and Critical Care Medicine, University of Tokyo Hospital, Tokyo, Japan; [3] Intensive Care Research, Austin Health, Melbourne, Victoria, Australia; [4] AM-Pharma B.V., Utrecht, The Netherlands; [5] Faculty of Medical Sciences, Radboud University, Nijmegen, The Netherlands

Abstract

Sepsis-associated AKI (SA-AKI) is associated with mortality and long-term sequelae, but no pharmacological options are available. Ilofotase alfa is a human recombinant alkaline phosphatase with renoprotective effects that showed improved renal function, survival and Major Adverse Kidney Events on day 90 (MAKE90) in SA-AKI patients in a Phase 2 study, STOP-AKI. 'REVIVAL', a large global phase 3 trial (NCT04411472), was conducted to confirm the efficacy and safety of Ilofotase alfa in patients with SA-AKI.

REVIVAL was conducted in SA-AKI patients (on vasopressor and <48hrs of AKI, according to KDIGO definition) recruited from 120 sites in Europe, North-America, Australia, New Zealand, and Japan. The primary endpoint was 28-day all-cause mortality. The key secondary endpoint was MAKE90. A pre-unblinding modified MAKE90 was defined as death up to Day 90, RRT through Day 28 and on Day 90, >25% drop in eGFR at Day 90, rehospitalization to Day 90.

The trial stopped for futility after 649 (Placebo n=319; IA n=330) patients, following a planned interim analysis with an observed 28-day mortality of 27.9% on both arms. No safety concerns were identified. A beneficial effect on the modified MAKE90 was observed (p=0.031), mainly driven by reduction of the number of patients receiving renal replacement therapy (RRT) (28.2% Ilofotase alfa vs 36.4% placebo). This effect was most pronounced in patients with pre-existent renal impairment.

Ilofotase alfa exerts renoprotective effects in SA-AKI. Further research is required to confirm a role for treatment of SA-AKI in patients with pre-existent renal impairment.

Introduction

Sepsis is the leading cause of acute kidney injury (AKI) in critically ill patients. Patients with SA-AKI are at risk of developing chronic kidney disease (CKD), resulting in a huge burden for patients and society. On the other hand, CKD is one of the risk factors for AKI. Several studies have shown that underlying CKD markedly increases the risk of AKI and that the risk increases proportionally with increasing CKD stage. Accordingly, CKD complicated by AKI in critically ill patients is common and leads to worse outcomes and delayed recovery of patients in the ICU. Currently, there are no medicinal products approved specifically for the treatment of SA-AKI, only supportive care such as fluid management and RRT is provided.

Ilofotase alfa is a human recombinant alkaline phosphatase, capable of dephosphorylating multiple damage-associated molecular pattern molecules such as ATP and pathogen associated molecular pattern molecules such as LPS, and through this mechanism it is thought to reduce systemic and local inflammation. In particular, the kidney is negatively affected by increased levels of ATP while adenosine has in tissue protective effects in renal function.

Previous clinical studies have demonstrated that patients treated with ilofotase alfa or bovine form of alkaline phosphatase, have better outcomes, including renal parameters and mortality, with a favorable tolerability profile compared to placebo.

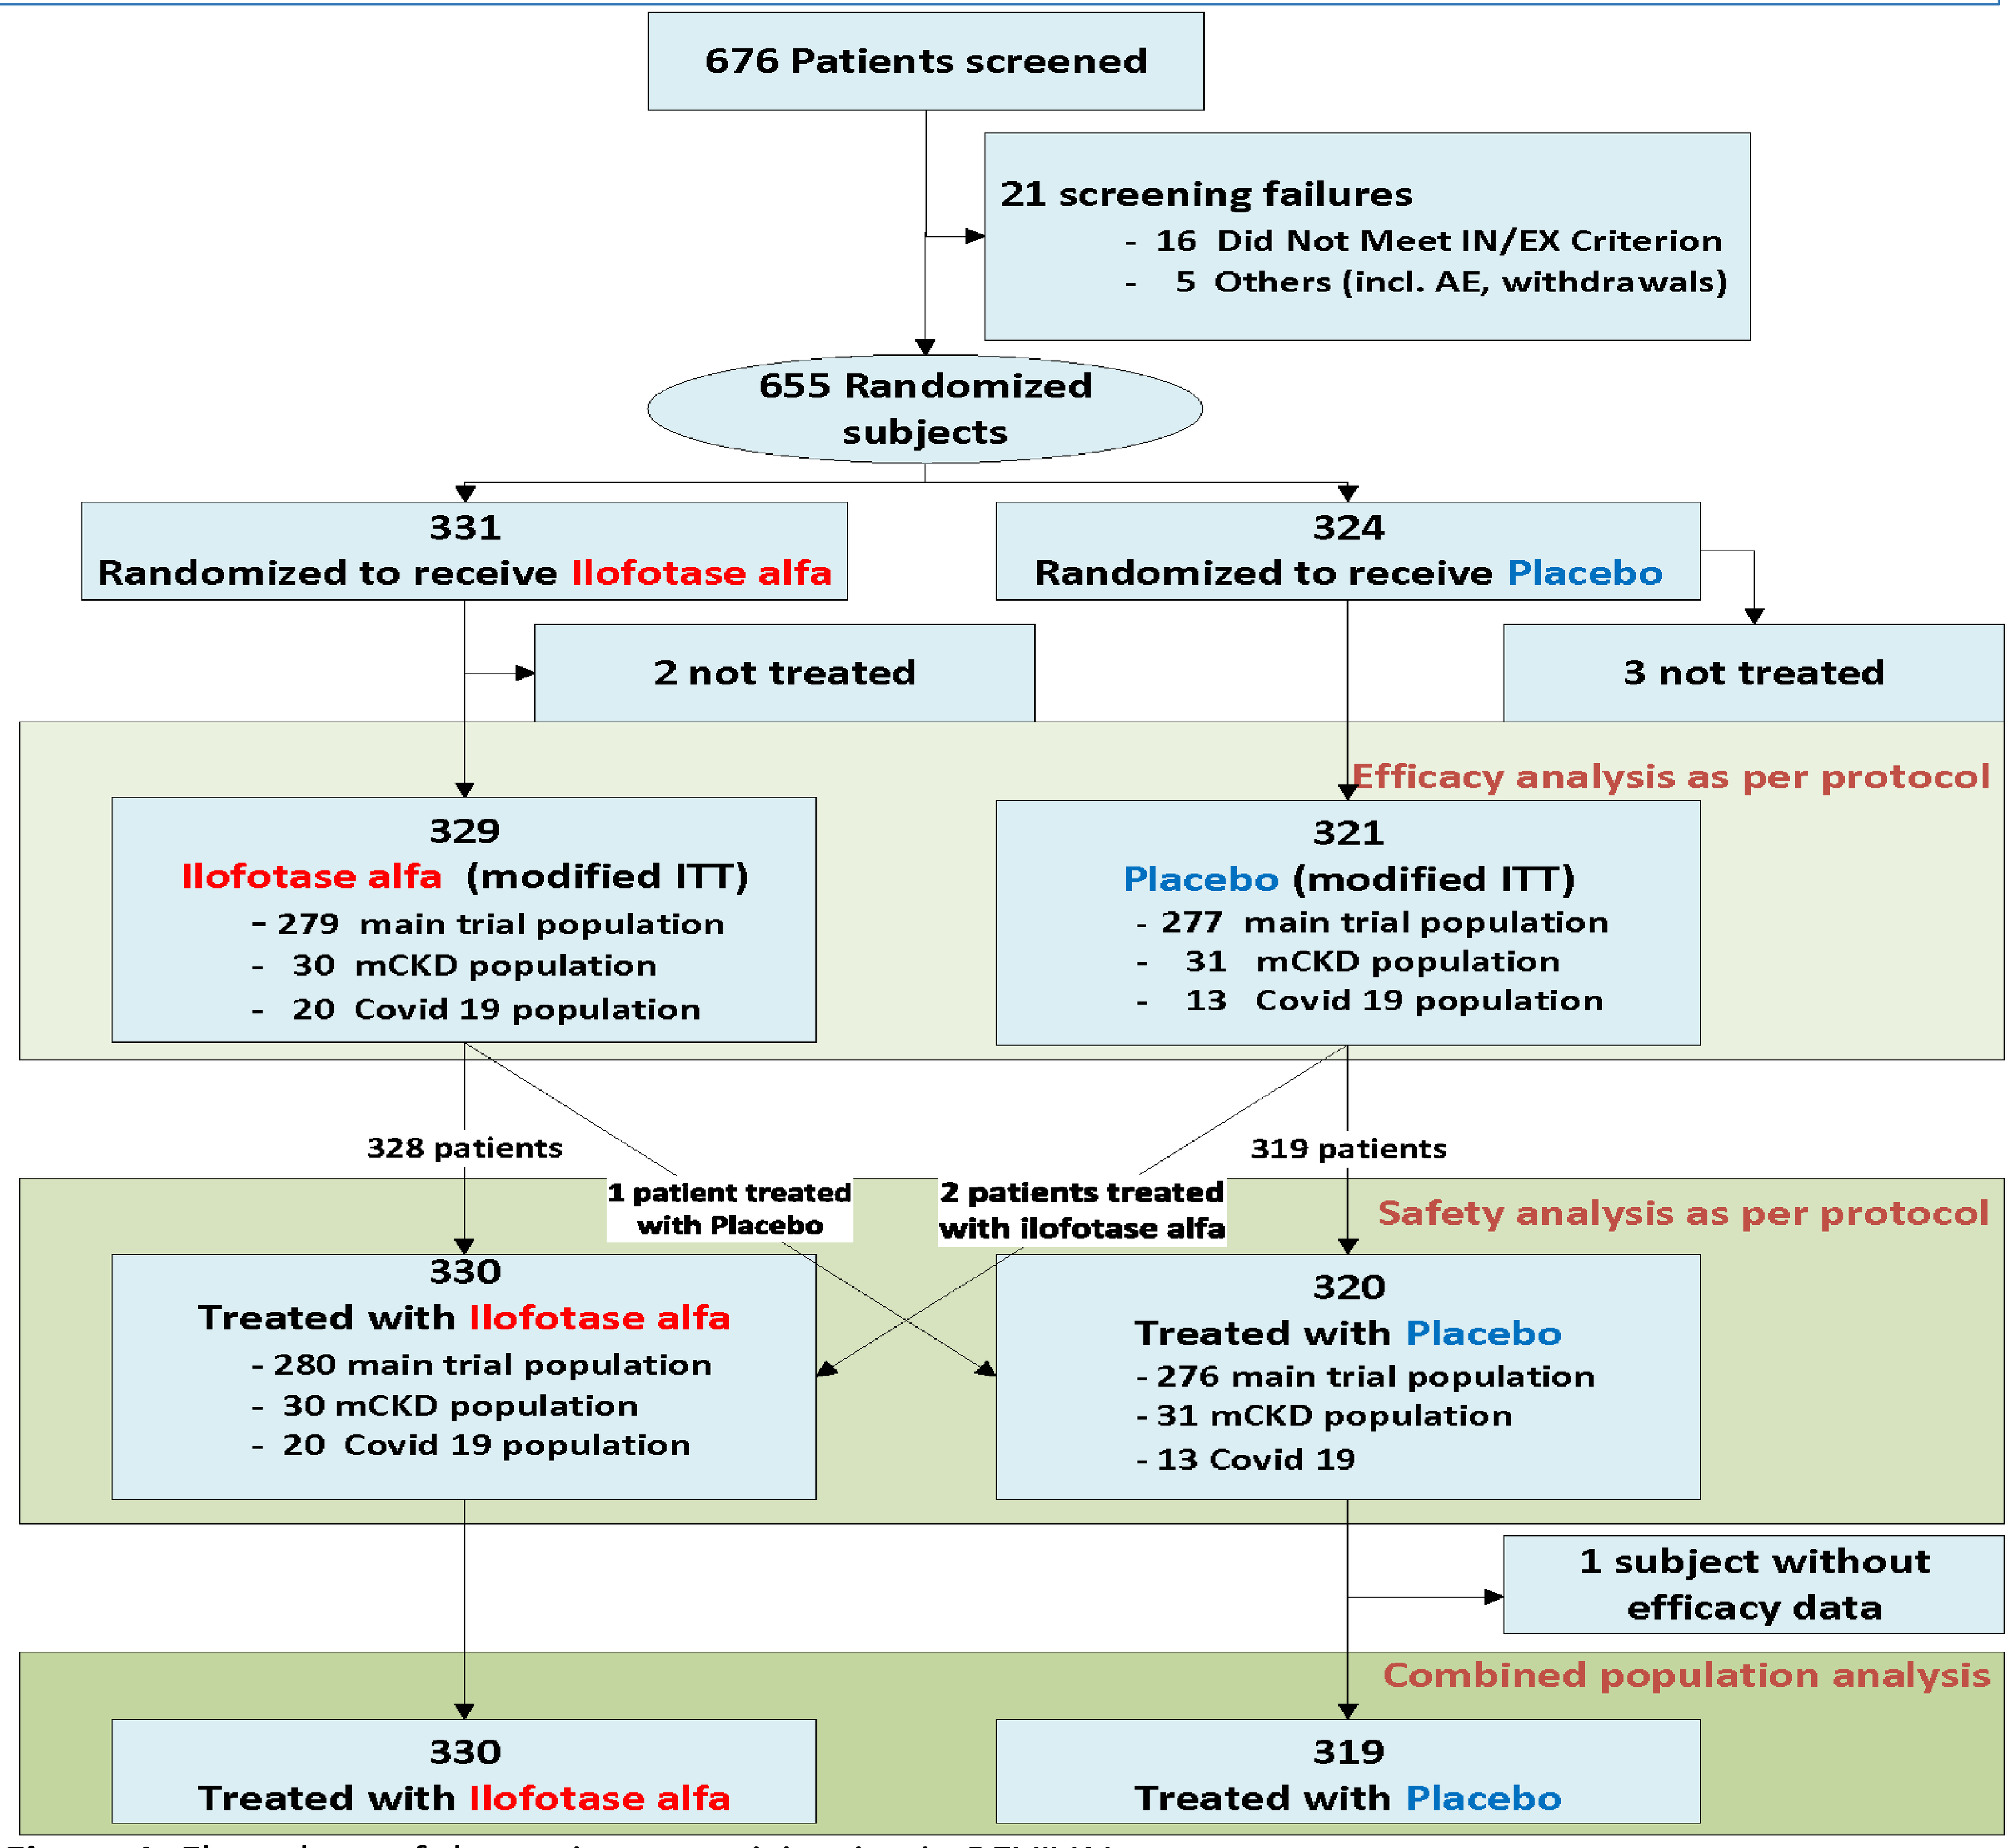
676 Patients screened
21 screening failures
- 16 Did Not Meet IN/EX Criterion
- 5 Others (incl. AE, withdrawals)
655 Randomized subjects
331 Randomized to receive Ilofotase alfa
324 Randomized to receive Placebo
2 not treated
3 not treated
Efficacy analysis as per protocol
329 Ilofotase alfa (modified ITT)
- 279 main trial population
- 30 mCKD population
- 20 Covid 19 population
321 Placebo (modified ITT)
- 277 main trial population
- 31 mCKD population
- 13 Covid 19 population
328 patients
1 patient treated with Placebo
2 patients treated with ilofotase alfa
319 patients
Safety analysis as per protocol
330 Treated with Ilofotase alfa
- 280 main trial population
- 30 mCKD population
- 20 Covid 19 population
320 Treated with Placebo
- 276 main trial population
- 31 mCKD population
- 13 Covid 19
1 subject without efficacy data
Combined population analysis
330 Treated with Ilofotase alfa
319 Treated with Placebo

Figure 1. Flow chart of the patients participating in REVIVAL

Materials and Methods

SA-AKI patients (on vasopressor and <48hrs of AKI, according to KDIGO definition) were recruited from 120 sites in Europe, North-America, Australia, New Zealand, and Japan. The primary endpoint was 28-day all-cause mortality. The key secondary endpoint was Major Adverse Kidney Events on day 90 (MAKE90). A pre-unblinding modified MAKE90 was defined as death up to Day 90, RRT through Day 28 and on Day 90, >25% drop in eGFR at Day 90, rehospitalization to Day 90.

		Combined patient population					
		Main Trial Population		'Moderate-to-Severe' CKD Population		COVID19 Population	
		ilofotase alfa (N=279)	Placebo (N=277)	ilofotase alfa (N=30)	Placebo (N=31)	ilofotase alfa (N=20)	Placebo (N=13)
Sex: n(%)	Female	105 (37.6%)	104 (37.5%)	12 (40.0%)	8 (25.8%)	6 (30.0 %)	2 (15.4 %)
Race: n(%)	AI or NA	1 (0.4%)	0	0	0	0	0
	Asian	16 (5.7%)	23 (8.3%)	0	0	1 (5.0 %)	1 (7.7 %)
	Black	6 (2.2%)	9 (3.2%)	1 (3.3%)	0	3 (15.0 %)	0
	Caucasian	233 (83.5%)	227 (81.9%)	26 (86.7%)	27 (87.1%)	15 (75.0 %)	11 (84.6 %)
	Multiple	1 (0.4%)	1 (0.4%)	0	0	0	0
	Other	2 (0.7%)	1 (0.4%)0	0	2 (6.5%)	1 (5.0 %)	1 (7.7 %)
	Not reported	20 (7.2%)	16 (5.8%)	2 (6.5%)	3 (10.0%)		
	Missing	0	1 (0.4%)	0	0	0	0
Age in years:		70.0 (62.0, 76.0)	69.0 (61.0, 76.0)	69.0 (66.0, 78.0)	75.0 (68.0, 79.0)	65.0 (54.5,71.5)	67.0 (64.0,75.0)
BMI in kg/m^2:		29.05 (25.00, 33.30)	27.09 (24.51, 34.14)	29.05 (24.49, 33.08)	29.20 (24.73, 32.66)	28.1 (24.7,32.1)	31.4 (28.3,35.0)
Pre AKI eGFR		75.63 (62.09, 89.98)	74.65 (62.73, 89.86)	39.63 (37.20, 78.00)	42.96 (35.77, 63.99)	72.8 (65.6,86.6)	74.2 (64.2,87.5)
AKI Diagnosis eGFR		31.48 (21.54, 43.25)	31.10 (20.40, 43.69)	22.85 (19.81, 42.12)	23.34 (19.17, 33.49)	32.7 (26.2,44.3)	32.5 (30.6,38.9)
mSOFA:		9.0 (7.0, 11.0)	9.0 (7.5, 11.0)	9.0 (8.0, 10.0)	9.0 (9.0, 10.0)	10.0 (8.0,11.0)	9.0 (8.0,10.0)
APACHE II Score:		23.0 (18.0, 28.0)	23.0 (18.0, 28.0)	23.0 (21.0, 27.0)	24.0 (22.0, 30.0)	23.0 (19.0, 30.0)	23.0 (18.0, 28.0)
MV Status: n(%)	On	209 (74.9%)	209 (75.5%)	20 (66.7%)	25 (80.6%)	18 (90.0 %)	13 (100%)
P/F ratio:		232.1 (156.0, 308.0)	206.0 (135.1, 306.1)	194.7 (105.0, 272.0)	178.4 (141.8, 220.9)	134.6 (97.1,195.0)	114.0 (97.0,146.6)
AKI stage: n(%)	0	11 (3.9%)	6 (2.2%)	2 (6.7%)	3 (9.7%)	10 (50.0 %)	8 (61.5 %)
	1	97 (34.8%)	100 (36.1%)	23 (76.7%)	13 (41.9%)		
	2	81 (29.0%)	95 (34.3%)	4 (13.3%)	9 (29.0%)	6 (30.0 %)	3 (23.1 %)
	3	90 (32.3%)	75 (27.1%)	1 (3.3%)	6 (19.4%)	4 (20.0 %)	2 (15.4 %)
	Missing	0	1 (0.4%)	0	0	0	0
KDIGO CKD stage	1	69 (24.7%)	67 (24.2%)	2 (6.7%)	4 (12.9%)	3 (15.0%)	3 (23.1%)
	2	153 (54.8%)	152 (54.9%)	9 (30.0%)	6 (19.4%)	13 (65.0%)	8 (61.5%)
	3a	50 (17.9%)	53 (19.1%)	0	2 (6.5%)	3 (15.0%)	2 (15.4%)
	3b	7 (2.5%)	3 (1.1%)	17 (56.7%)	15 (48.4%)		0
	4	0	1 (0.4%)	2 (6.7%)	3 (9.7%)	1 (5.0%)	0
	5	0	0	0	1 (3.2%)	0	0
	missing	0	1 (0.4%)		0	0	0

Table 1. Demographic and baseline characteristics by analysis population and treatment received

Results

Mortality up to day 90 for the combined trial populations is presented in Figure 2. Following Kaplan-Meier analysis in the combined patient population there was no statistical difference in mortality between the ilofotase alfa and the placebo groups neither at Day 28 (27.97% vs 28.12% respectively, p=0.48) nor at Day 90 (34.46% vs 36.03% respectively, p=0.34).

For **renal endpoints** Kaplan-Meier analysis of MAKE90, defined as death by day 90, OR eGFR decrease >25% at day 28 AND day 90 OR on RRT on day 90, was not statistically different between the ilofotase alfa and placebo groups in the combined population. MAKE90, defined as death by day 90, OR eGFR decrease >25% at day 90, OR rehospitalization, OR on RRT through day 28 or on day 90, was analyzed using the Kaplan-Meier method for the combined population group (Figure 3A). Ilofotase alfa demonstrated a significantly lower proportion of patients (57.2%) experiencing MAKE90 events compared with patients receiving placebo (64.7%) (p=0.031) . This effect was mainly driven by the component 'RRT through Day 28 OR on Day 90'. There was interaction between pre-AKI eGFR and the risk for MAKE90. The therapeutic efficacy of ilofotase alfa was more pronounced in patients with a lower pre-AKI eGFR (p=0.0235). (Figure 3B).

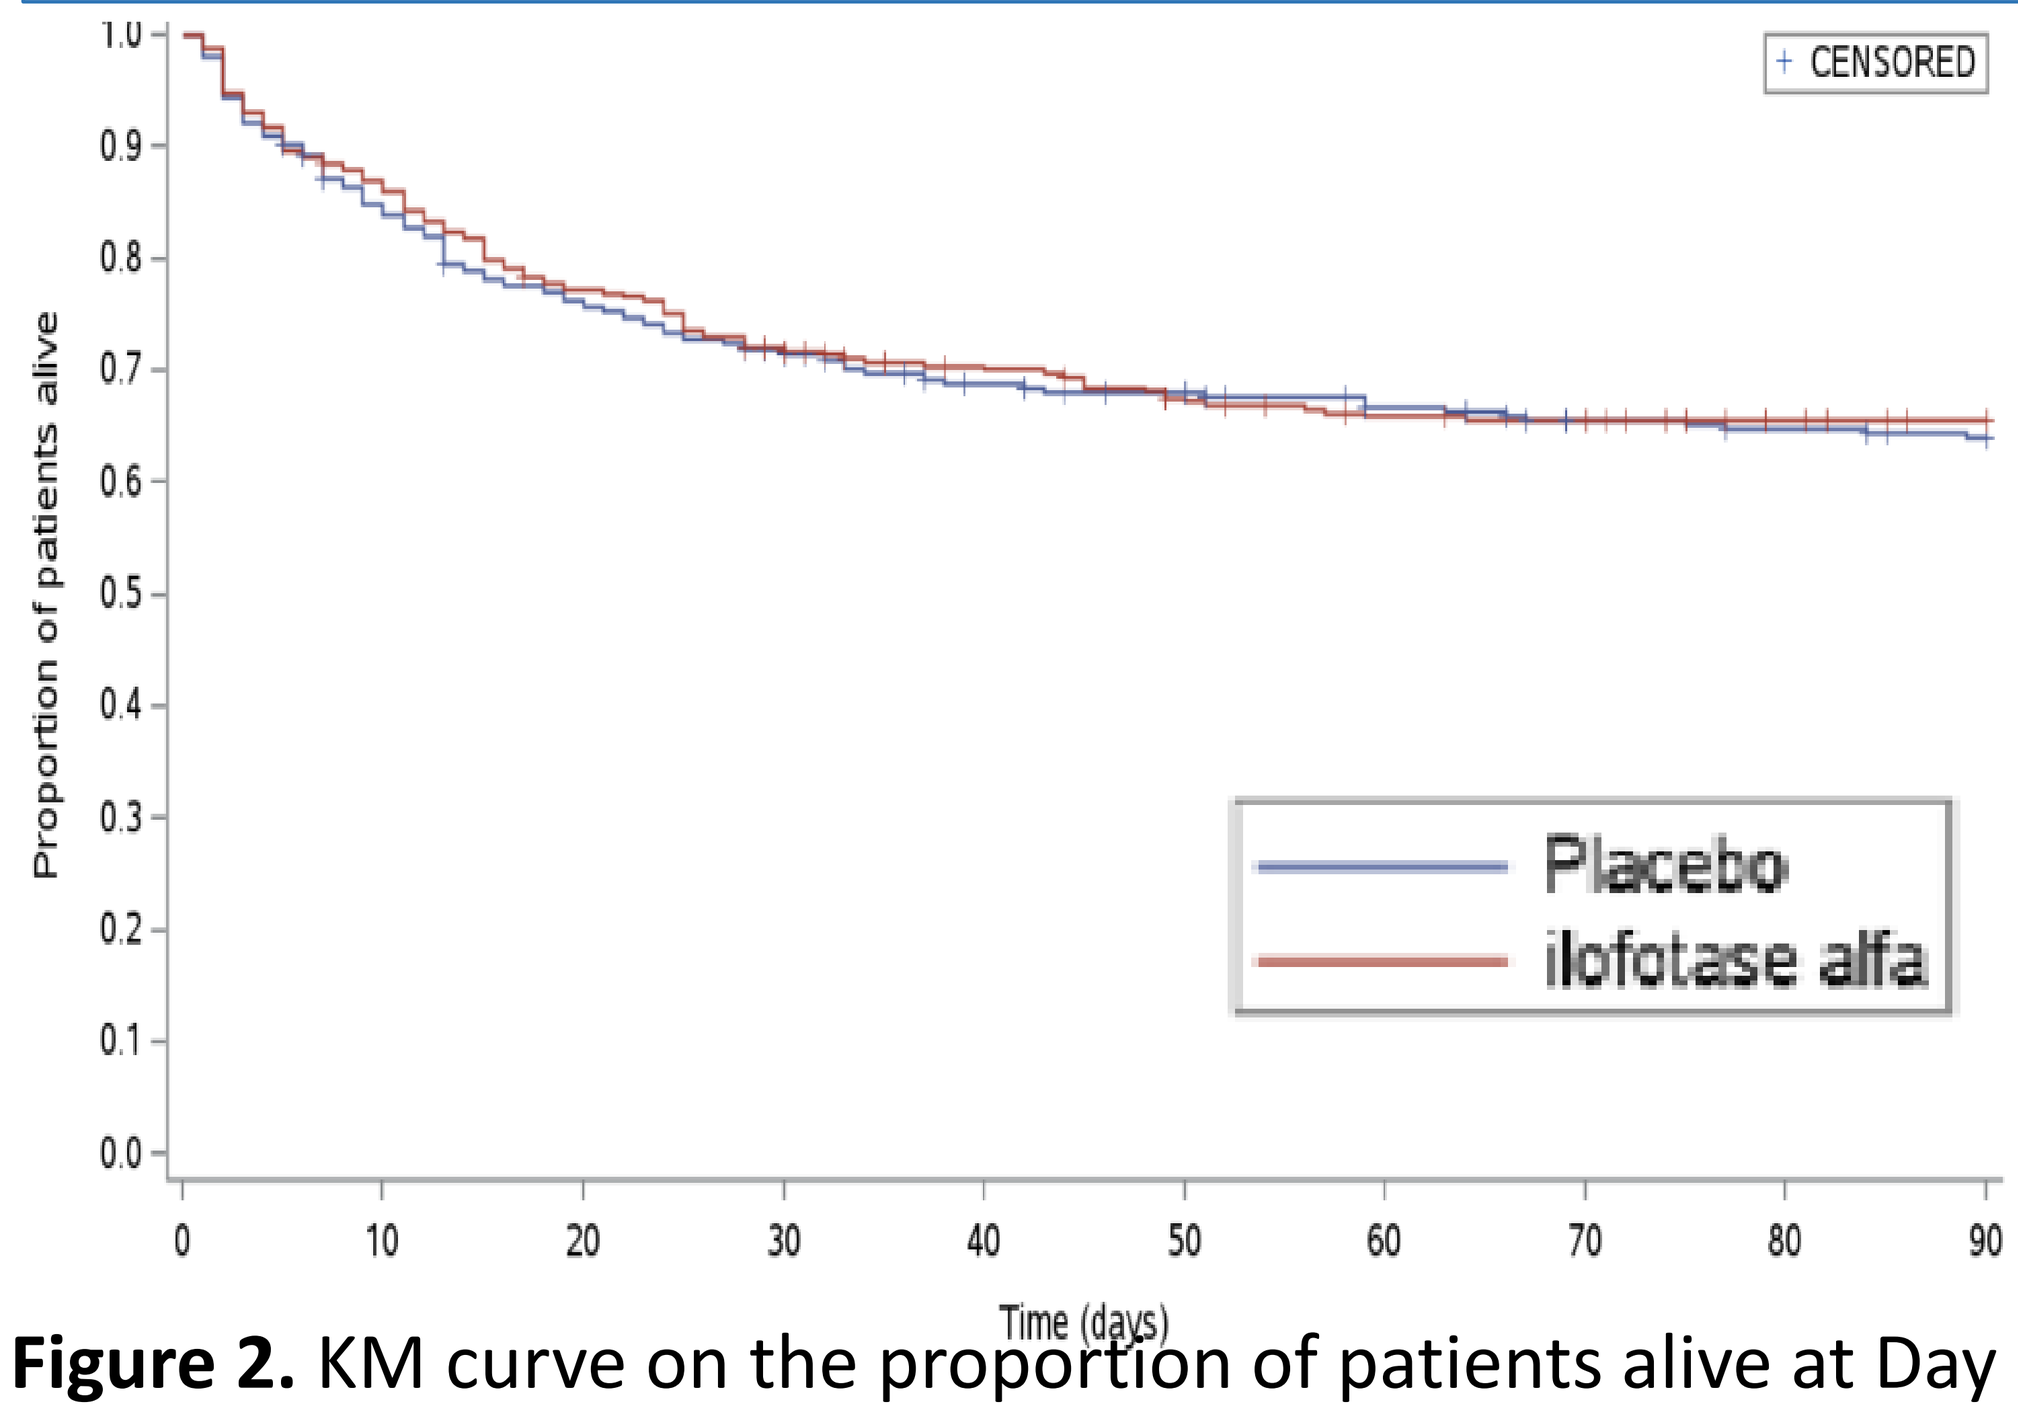
+ CENSORED
Proportion of patients alive
1.0
0.9
0.8
0.7
0.6
0.5
0.4
0.3
0.2
0.1
0.0
0
10
20
30
40
50
60
70
80
90
Time (days)
Placebo
ilofotase alfa

Figure 2. KM curve on the proportion of patients alive at Day 90 in the combined population.

Conclusions

- No confirmation of previously demonstrated survival benefit
- Renal protective effect observed which is consistent with previous trials
- A new trial focused on patients with pre-existent impaired renal function will be necessary to confirm the more pronounced therapeutic efficacy in these patients

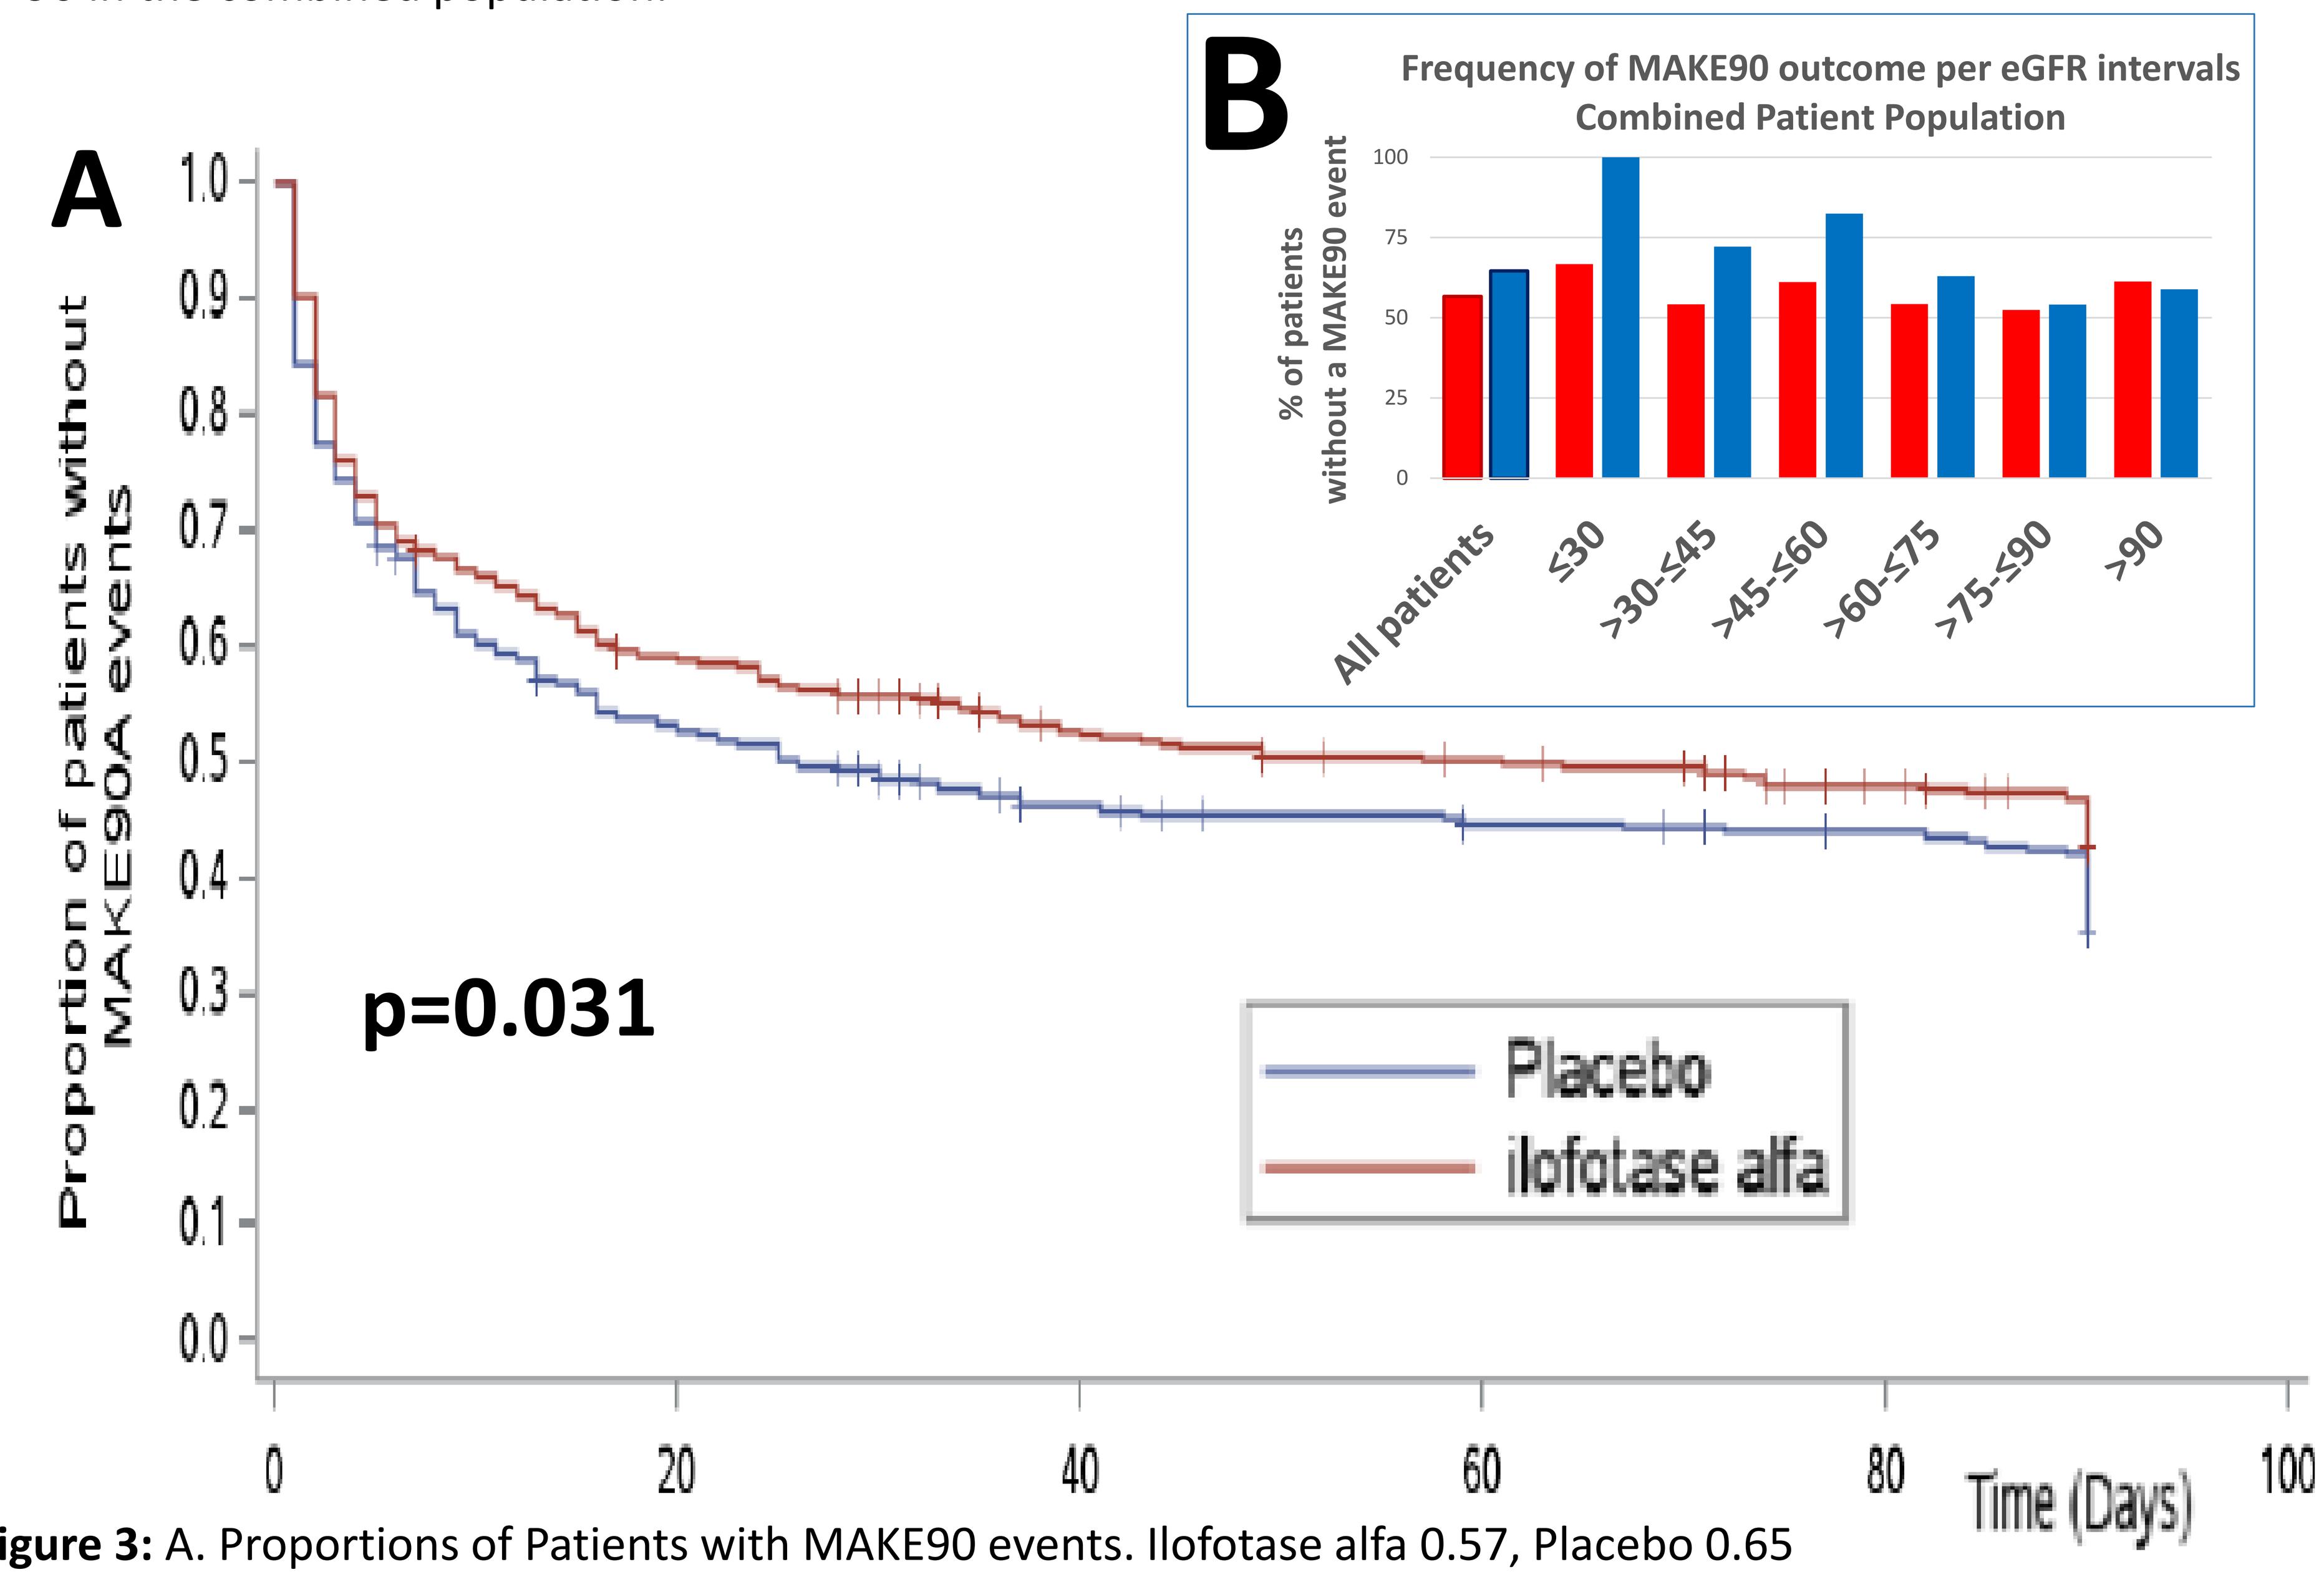
A
Proportion of patients without MAKE90A events
1.0
0.9
0.8
0.7
0.6
0.5
0.4
0.3
0.2
0.1
0.0
0
20
40
60
80
100
Time (Days)
p=0.031
Placebo
ilofotase alfa
B
Frequency of MAKE90 outcome per eGFR intervals
Combined Patient Population
% of patients without a MAKE90 event
100
75
50
25
0
All patients
≤30
>30-≤45
>45-≤60
>60-≤75
>75-≤90
>90

Figure 3: A. Proportions of Patients with MAKE90 events. Ilofotase alfa 0.57, Placebo 0.65
B. Frequency of MAKE90 outcomes per eGFR interval. The renal therapeutic efficacy of ilofotase alfa is more pronounced in patients with a lower pre-AKI eGFR (**p=0.0235**)

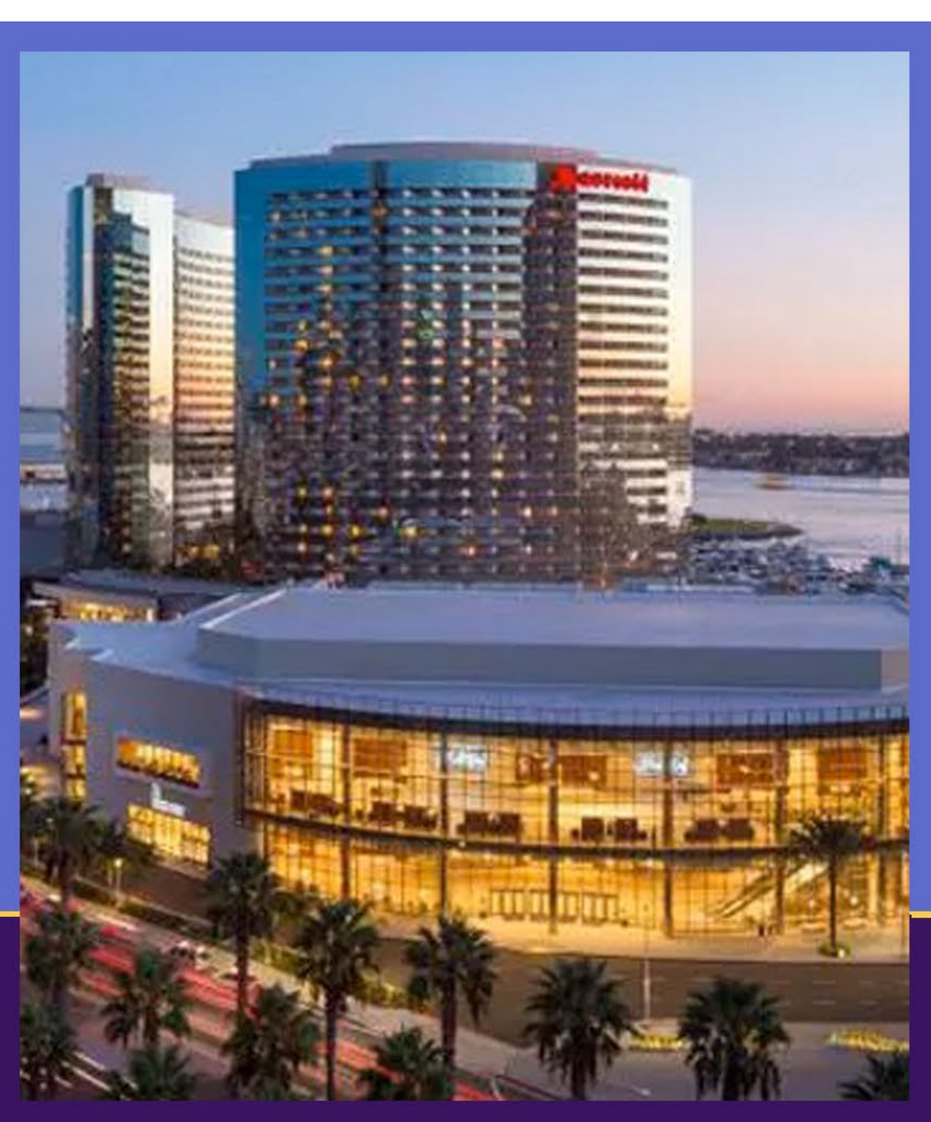

THE 28TH INTERNATIONAL CONFERENCE ON
ADVANCES IN CRITICAL CARE NEPHROLOGY
AKI & CRRT 2023
MARCH 29 - APRIL 1 SAN DIEGO, CALIFORNIA